Efficacy and Safety of 8 Weeks of Glecaprevir/Pibrentasvir in Treatment-Naïve Adults With HCV Genotype 1–6 and Aspartate Aminotransferase to Platelet Ratio Index (APRI) ≤1

FRI-653

Robert J Fontana[1], Sabela Lens[2], Stuart McPherson[3], Magdy Elkhashab[4], Krum Katzarov[5], Ana Gabriela Pires dos Santos[6], Zhenyi Xue[6], Roger Trinh[6], Lino Rodrigues Junior[6], Ariel Porcalla[6], Stefan Zeuzem[7]

[1]Division of Gastroenterology and Hepatology, University of Michigan, Ann Arbor, Michigan, United States; [2]Liver Unit, Hospital Clinic, IDIBAPS and CIBERehd, Barcelona, Spain; [3]Liver Unit, The Newcastle upon Tyne Hospitals NHS Foundation and Trust and Newcastle University, United Kingdom; [4]Toronto Liver Centre, Toronto, Ontario, Canada; [5]Department of Gastroenterology, Hepato-Pancreato-Biliary (HPB) Surgery and Transplantology, Military Medical Academy, Sofia, Bulgaria; [6]AbbVie Inc., North Chicago, Illinois, United States; [7]JW Goethe University, Frankfurt, Germany

Presented at The Liver Meeting of the American Association for the Study of Liver Diseases (AASLD), November 9–13, 2018, San Francisco, California, United States

INTRODUCTION

- WHO global target of HCV elimination by 2030 depends on access to testing and treatment with direct acting antivirals[1]
- Linkage to care following HCV diagnosis is a gap in the HCV care cascade, in part due to need for specialists in the comprehensive and costly evaluation for presence or absence of cirrhosis using either liver biopsy or transient elastography[1,2]
- Aspartate aminotransferase to platelet ratio index (APRI) is a low-cost, widely available, non-invasive method, for which APRI ≤1 has high negative predictive value (94%) for cirrhosis compared to biopsy[3,4]
- Non-invasive markers, including APRI, may be used to assess for cirrhosis prior to HCV treatment according to WHO, EASL, Australian, and Canadian guidelines[5–8]

G/P is Approved for Patients With HCV GT1–6 Infection

Glecaprevir pangenotypic NS3/4A protease inhibitor

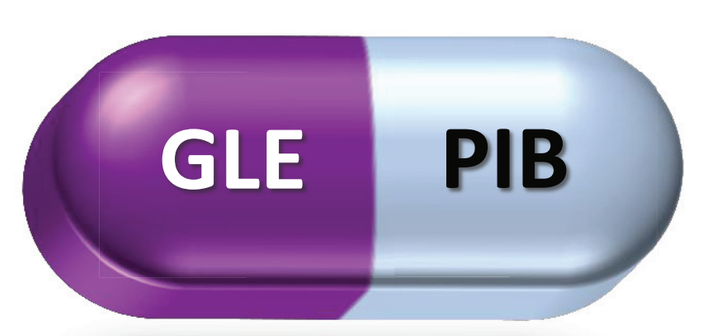
GLE
PIB
Coformulated: G/P

Pibrentasvir pangenotypic NS5A inhibitor

- 8-week duration approved for treatment-naïve patients with HCV genotype (GT) 1–6 infection and without cirrhosis[9]
- Pangenotypic SVR12 rate of 98% in more than 2200 patients in registrational studies
- Potent against common polymorphisms (eg, Y93H in NS5A and Q80K in NS3)
- Favorable safety profile irrespective of baseline factors
- Recent real-world results demonstrate that G/P achieved high SVR12 rates consistent with those observed in clinical trials[10,11]

G/P dosed as 3 pills taken once daily with food for a total dose of 300 mg/120 mg.
Glecaprevir was identified by AbbVie and Enanta.

OBJECTIVE

Evaluate whether treatment-naïve patients with APRI ≤1 would achieve high SVR12 rates with 8-week G/P regimen.

METHODS

- Phase 3, open label, single-arm, multicenter study conducted from August 7th, 2017 to August 13th, 2018
 - Patients were enrolled in Bulgaria, Canada, France, Germany, Poland, Puerto Rico, Russia, Spain, United Kingdom, and United States of America
- Data were analyzed on an intent-to-treat (ITT) population (all patients who received ≥1 dose of G/P) and on a modified ITT (mITT) population (excluding patients who did not achieve SVR12 for reasons other than virologic failure, like missing SVR12 data or premature G/P discontinuation)
 - Virologic failure included any virologic breakthrough during treatment or post-treatment relapse
- APRI was assessed at screening using the following formula:

$$APRI = \left(\frac{\frac{AST\,(\frac{U}{L})}{AST\,(ULN)\,(\frac{U}{L})}}{Platelet\ Count\,(\frac{10^9}{L})} \right) \times 100$$

METHODS (CONTINUED)

Figure 1. Study Design

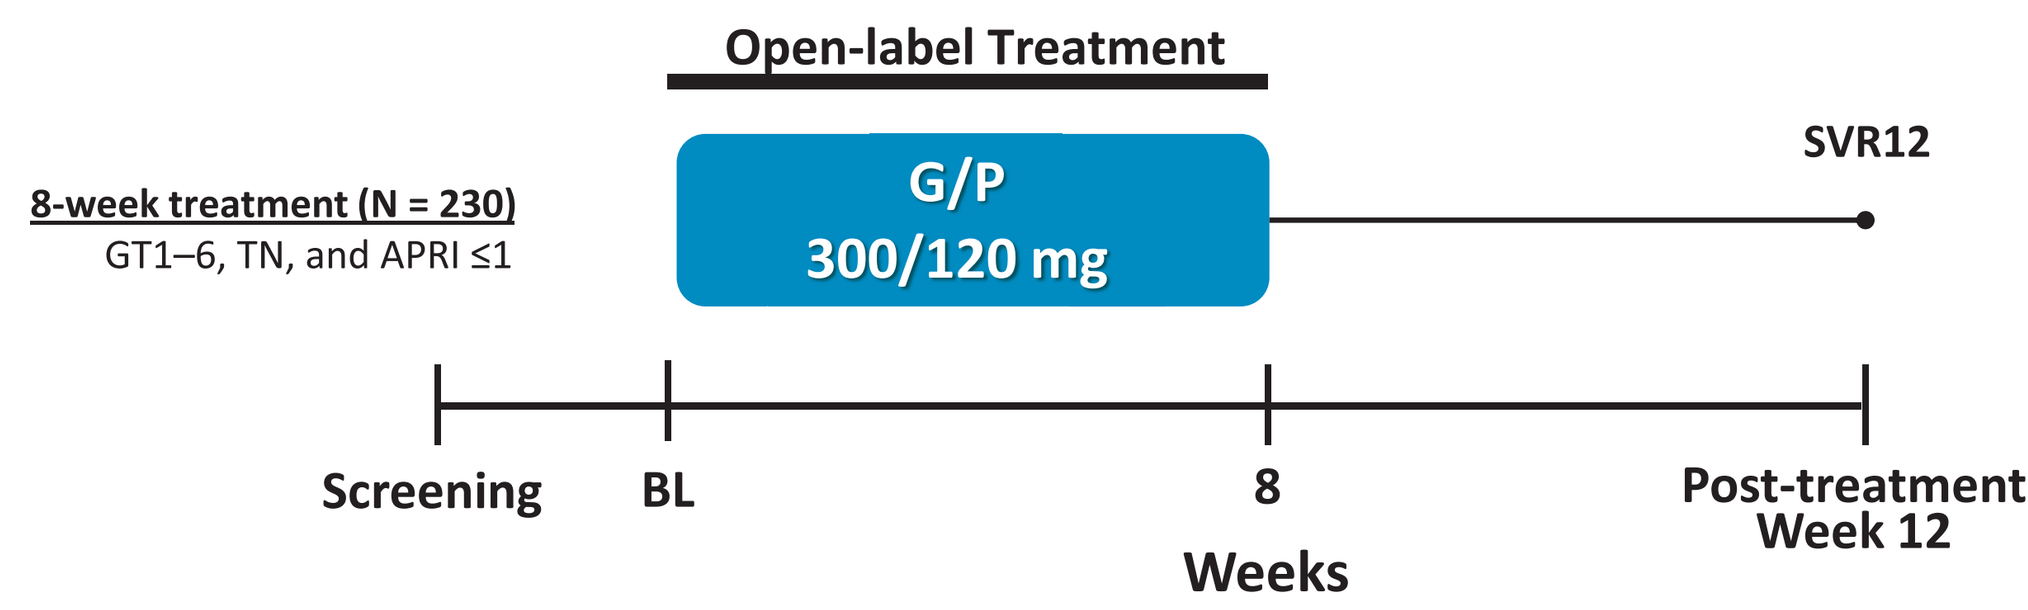
Open-label Treatment
8-week treatment (N = 230)
GT1–6, TN, and APRI ≤1
G/P
300/120 mg
SVR12
Screening
BL
8
Post-treatment Week 12
Weeks

BL, baseline; G/P, glecaprevir/pibrentasvir; SVR12, sustained virologic response at post-treatment Week 12; TN, treatment-naïve; APRI, aspartate aminotransferase to platelet ratio index; GT, genotype.

KEY INCLUSION CRITERIA

- Adult male or female, at least 18 years old at the time of screening
- HCV treatment naïve with chronic HCV genotype (GT) 1, 2, 3, 4, 5, or 6 infection (mixed GT and indeterminate GT were acceptable)
- Positive anti-HCV Ab and HCV RNA ≥1000 IU/mL for ≥6 months prior to and at screening
- APRI score ≤1 at the time of screening
- HIV-1 negative or HIV-1 positive (naïve or on stable qualifying antiretroviral therapy)

KEY EXCLUSION CRITERIA

- Any prior evidence of cirrhosis; however, formal assessment for cirrhosis was not required
- Presence of hepatitis B or hepatocellular carcinoma
- ALT or AST >10 × upper limit of normal (ULN)
- Direct bilirubin >ULN
- Platelet count <150 000 cells/mm^3
- Albumin <lower limit of normal
- Creatinine clearance (Cockcroft-Gault) <30 mL/min
- History of organ transplantation

ENDPOINTS AND ANALYSES

Sequential Efficacy Endpoints (fixed sequence testing procedure):

Primary Efficacy Endpoint
Percentage of patients with SVR12 (HCV RNA <LLOQ 12 weeks after the last dose of study drug) in the mITT population
- Efficacy will be established if the lower bound of the 95% confidence interval (95%CI) is greater than the threshold of 92.4%, based on the historical rate observed in GLE/PIB registrational studies in treatment-naïve, non-cirrhotic subjects (98.4%) minus a margin of 6%

Secondary Efficacy Endpoints
Percentage of patients with SVR12 in the ITT population
- Efficacy will be established if the lower bound of the 95%CI is greater than the threshold of 91.4%, based on the mITT threshold minus an expected 1% rate of non-virological SVR failures

Non-sequential Endpoints:

Secondary Efficacy Endpoints
- Percentage of patients with on-treatment virologic failure in ITT population
- Percentage of patients with post-treatment relapse in ITT population

Additional Assessments:

Additional Analyses
- Treatment adherence* conducted in ITT population
- Number and percentage of patients with viral suppression (HCV RNA <LLOQ) at treatment visits in patients with available data

Safety Assessments
Adverse events (AEs)† and key laboratory abnormalities were assessed in ITT population

SVR12, sustained virologic response at 12 week post-treatment; HCV, hepatitis C virus; LLOQ, lower limit of quantification; ITT, intent-to-treat; CI, confidence interval; mITT, modified ITT; AE, adverse event.
*Treatment adherence defined as use of ≥80% and ≤120% of the expected total number of tablets.
†Includes AEs leading to treatment discontinuations, AEs occurring in ≥5% of patients, serious AEs, and DAA-related serious AEs.

RESULTS

PATIENTS

- The Baseline Demographics and Disease Characteristics are summarized in **Table 1** for both the mITT and ITT populations
- Among the 230 patients in the ITT population, 99 (43%) had a history of a psychiatric disorder

Table 1. Baseline Demographics and Disease Characteristics

Characteristic	mITT population N = 222	ITT population N = 230
Male, n (%)	111 (50)	117 (51)
Race, n (%)		
White	202 (91)	207 (90)
Black or African American	10 (5)	13 (6)
Asian	10 (5)	10 (4)
Hispanic or Latino ethnic origin, n (%)	23 (10)	25 (11)
Age, median (range), years	48 (19–82)	48 (19–82)
Age ≥65 years old, n (%)	23 (10)	25 (11)
BMI, median (range), kg/m^2	25.3 (16. 9–55.6)	25.2 (16. 9–55.6)
Baseline HCV RNA level, median (range), log_{10} IU/mL	6.3 (2.2–7.7)	6.3 (2.2–7.7)
Baseline HCV RNA ≥1 million IU/mL, n (%)	141 (64)	146 (63)
HCV genotype, n (%)		
GT1	145 (65)	151 (66)
GT1a	77 (35)	82 (36)
GT1b	67 (30)	68 (30)
GT2	33 (15)	33 (14)
GT3	33 (15)	35 (15)
GT4	9 (4)	9 (4)
GT6	2 (<1)	2 (<1)
Key baseline polymorphisms, n (%)*		
Any NS3 polymorphism	1 (<1)	1 (<1)
Any NS5A polymorphism	68 (31)	71 (31)
Screening APRI, median (range)	0.41 (0.13–1.0)	0.41 (0.`1.0)
Screening APRI, n (%)		
≤0.5	136 (61)	140 (61)
0.5–0.7	54 (24)	57 (25)
0.71–1.0	32 (14)	33 (14)
Blinded Fibrotest, median (range)†	0.26 (0.02–0.87)	0.26 (0.02–0.87)
Platelet count, median (range), count/10^9/L	243 (126–462)	243 (126–483)
HIV co-infection, n (%)‡	8 (4)	10 (4)
CD4+ T-cell count, median (range), cells/mm^3§	762 (444–1199)	692 (444–1199)
History of injection drug use, n (%)	83 (37)	87 (38)
Within the last 12 months	5 (2)	6 (3)
More than 12 months ago	78 (35)	81 (35)
On stable opiate substitution, n (%)	19 (9)	19 (8)
History of diabetes, n (%)	7 (3)	9 (4)
History of depression or bipolar disorder, n (%)	43 (19)	45 (20)

BMI, body-mass index; HCV, hepatitis C virus; GT, genotype; APRI, Aspartate aminotransferase to platelet ratio; HIV, human immunodeficiency virus.
*Includes any baseline resistance-associated variants in NS3 (155, 156, and 168) or NS5A (24, 28, 30, 31, 58, 92, and 93) at a 15% detection threshold. No patients had both a NS3 and a NS5A key resistance-associated variant.
†Performed at baseline, blinded to the investigators and therefore, not used for patient eligibility.
‡All patients with HIV co-infection were antiretroviral therapy-naïve.
§Only assessed in the 10 HIV/HCV co-infected patients.

EFFICACY

- The primary efficacy endpoint was met, with an overall SVR12 rate in the mITT population of 100% (222/222, 95%CI = 98.3–100) **(Figure 2)**
- The sequential secondary endpoint was also met, with an overall SVR12 rate in the ITT population of 96.5% (222/230, 95%CI = 94.2–98.9). Eight (3%) patients did not achieve SVR12 due to non-virologic failure reasons **(Figure 2)**
- No patients experienced on-treatment virologic failure or post-treatment relapse
- High SVR12 rates were observed in all HCV genotypes **(Figure 3)**
- Nighty-nine percent (202/204) of patients were adherent to the treatment (≥80% and ≤120% adherence) among patients with available data at all treatment visits

Figure 2. Efficacy of 8-week G/P Regimen in HCV Treatment-naïve Patients With APRI ≤1

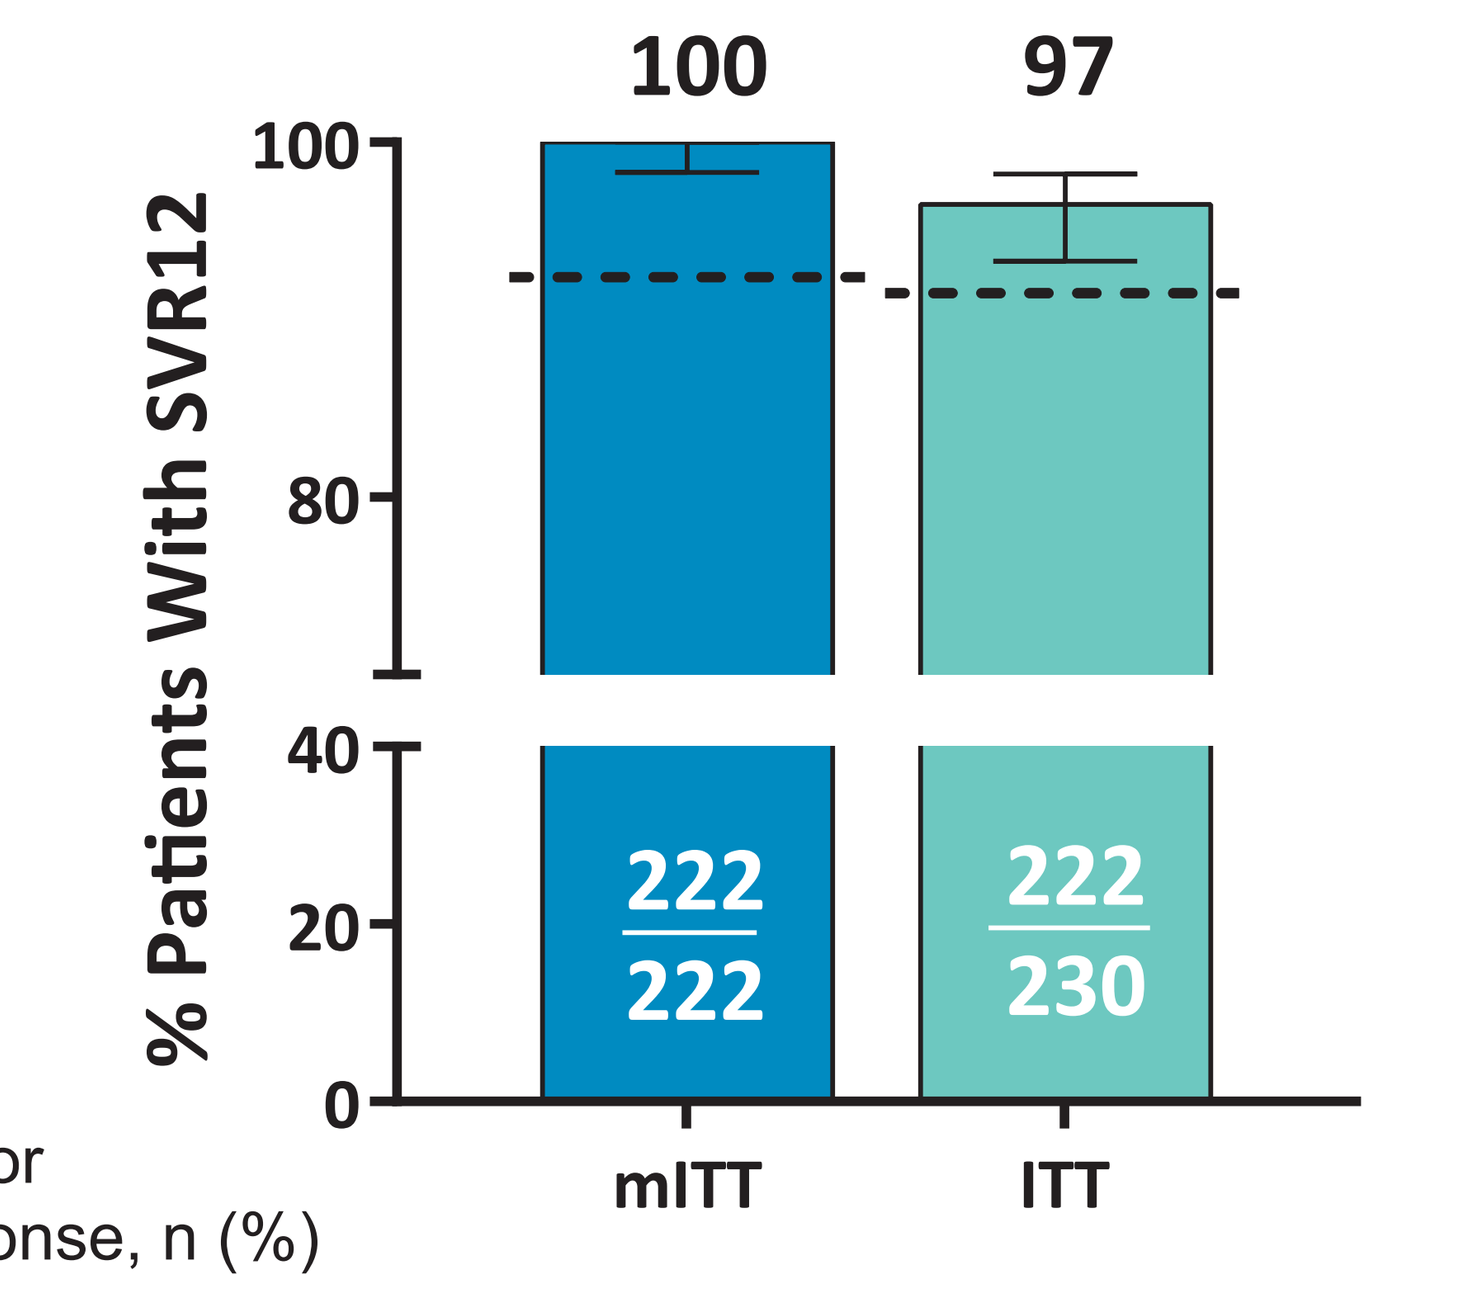
100
97
% Patients With SVR12
100
80
40
20
0
222
222
222
230
mITT
ITT

Reason for non-response, n (%)	mITT	ITT
Breakthrough	0	0
Relapse	0	0
Premature G/P d/c	N/A	3 (1%)*
Missing SVR_{12}	N/A	5 (2%)†

mITT, modified intent-to-treat; ITT, intent-to-treat; SVR12, sustained virologic response at 12-week post-treatment; CI, confidence interval; G/P, glecaprevir/pibrentasvir; d/c discontinuation.
Dotted lines indicate threshold above which lower bound of 95%CI must be greater than in mITT and ITT populations in order to meet primary and secondary endpoint, respectively.
*Includes 2 patients who prematurely discontinued due to a serious AE (see **Table 3**).
†All 5 patients missing SVR12 data had no detectable HCV RNA at end of treatment.

Table 2. Number and Percentage of Patients With Viral Suppression Over Time

Viral Suppression at Treatment Visit, n/N (%)*	8-week G/P treatment
Week 4	214/225 (95)
Week 8 (EOT)	211/211 (100)
Final treatment visit†	226/226 (100)

G/P, glecaprevir/pibrentasvir; EOT, end of treatment.
*Viral suppression reported as number and percentage of patients with HCV RNA less than lower limit of quantification at each post-baseline treatment visit in patients with available SVR data.
†The last visit for a patient during G/P treatment.

Figure 3. Efficacy by Genotype for 8-week G/P Regimen in HCV Treatment-naïve Patients With APRI ≤1

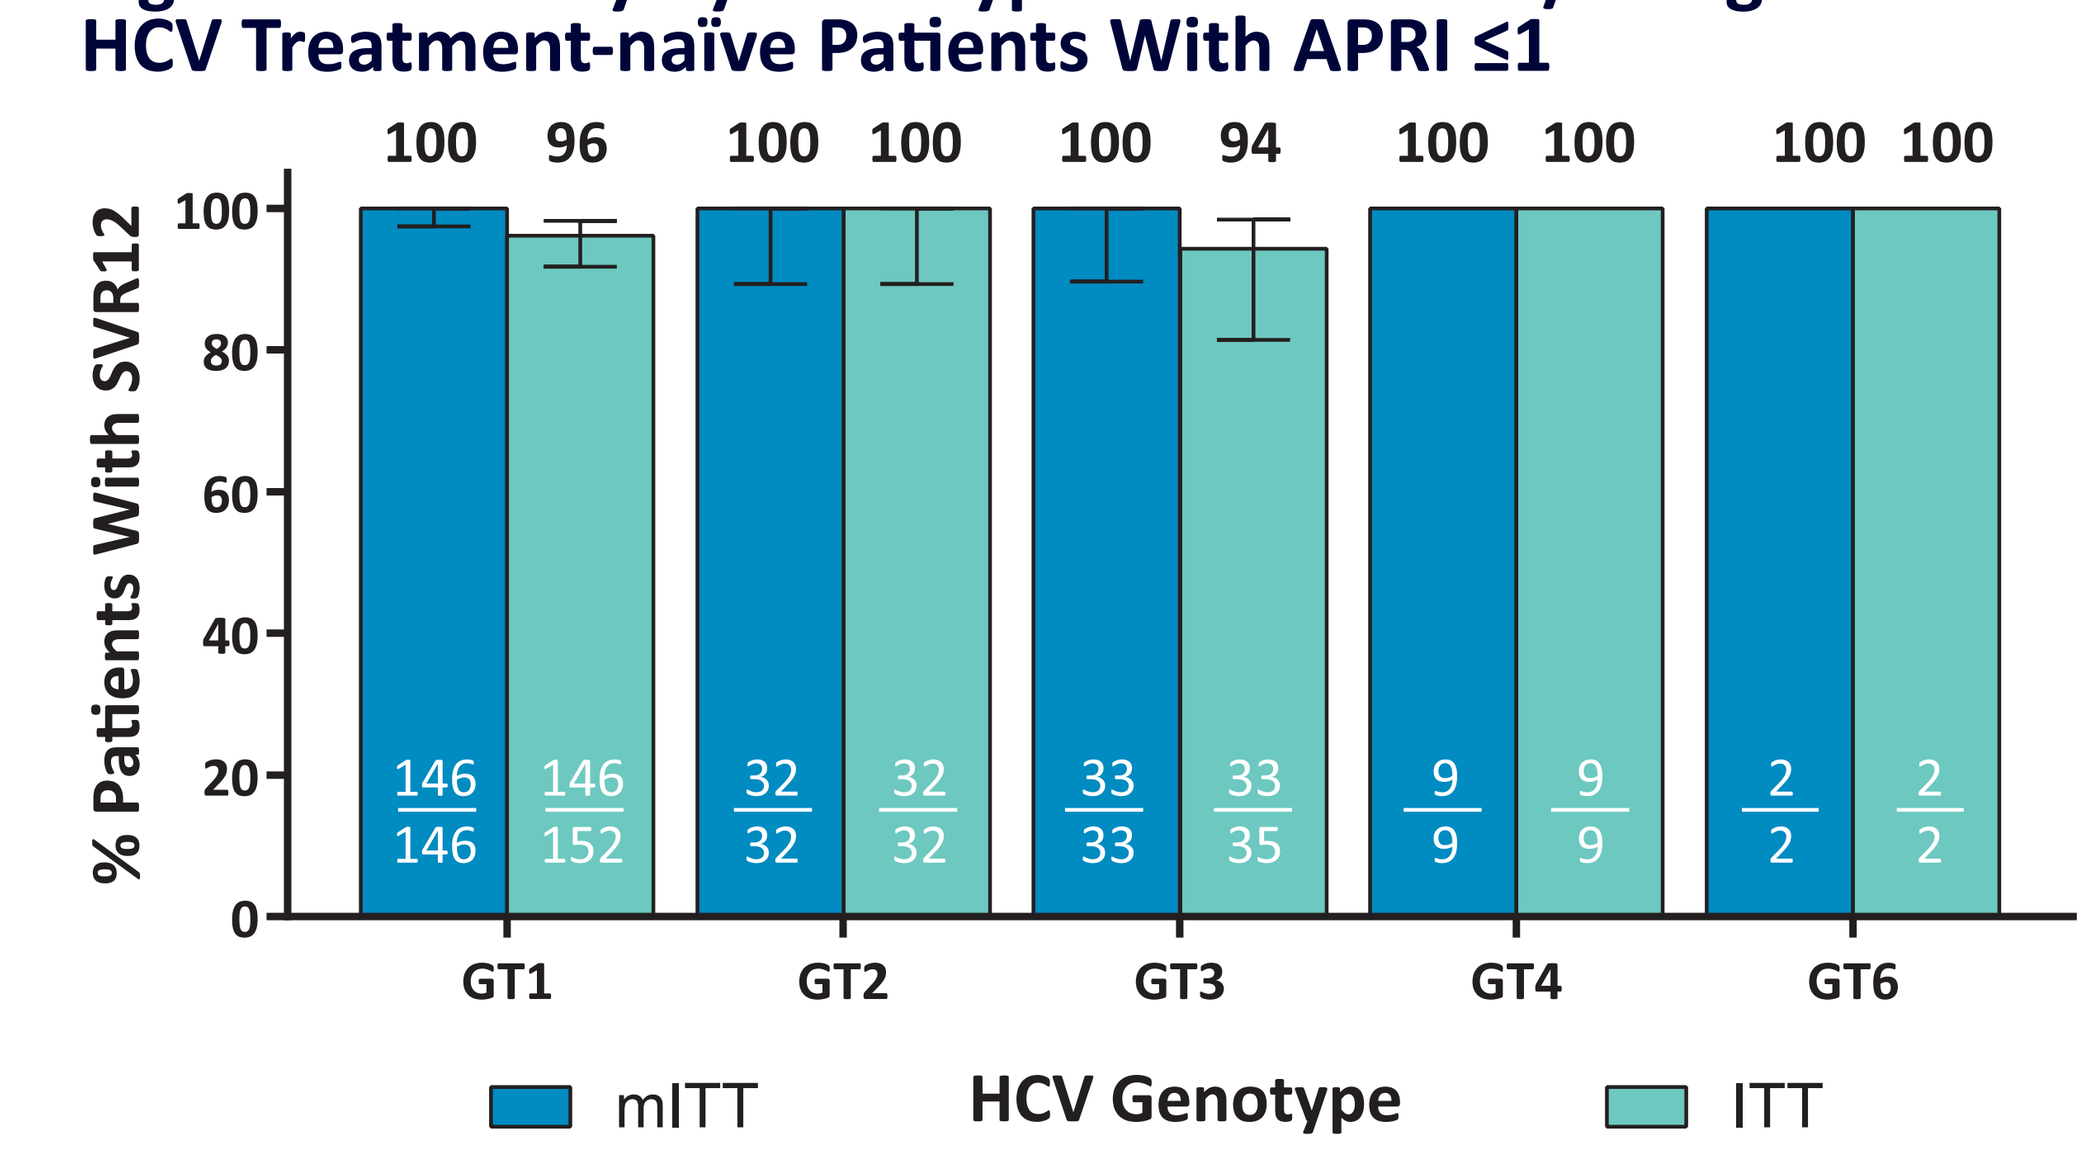
100
96
100
100
100
94
100
100
100
100
% Patients With SVR12
100
80
60
40
20
0
146/146
146/152
32/32
32/32
33/33
33/35
9/9
9/9
2/2
2/2
GT1
GT2
GT3
GT4
GT6
mITT
HCV Genotype
ITT

mITT, modified intent-to-treat; ITT, intent-to-treat; GT, genotype; SVR12, sustained virologic response at 12-week post-treatment; HCV, hepatitis C virus.

SAFETY

- Overall, 124 (54%) patients experienced an AE, of which 8 (3%) patients had a grade 3 or higher AE **(Table 3)**
- Two (1%) patients experienced serious AEs of angioedema that led to premature G/P discontinuation
 - Both were African American and former drug users (both risk factors for angioedema) and were taking an ACE inhibitor (lisinopril), a common cause of drug-induced angioedema[12]
- The most common AEs (occurring in ≥5% of all patients) were headache (13%) and fatigue (7%)
- No grade ≥3 laboratory abnormalities in ALT, AST, or total bilirubin were observed

Table 3. Adverse Events and Laboratory Abnormalities

Event, n (%)	ITT population N = 230
Any AE	124 (54)
Grade ≥3 AE	8 (3)
Serious AE	4 (2)
DAA-related* serious AE	2 (1)†
AE leading to premature G/P discontinuation	2 (1)†
AEs occurring in ≥5% of all patients by preferred term	
Headache	29 (13)
Fatigue	17 (7)
Laboratory abnormalities (Grade ≥3)	
ALT >5 × ULN‡	0
AST >5 × ULN	0
Total bilirubin >3 × ULN	0
Deaths	0

G/P, glecaprevir/pibrentasvir; AE, adverse event; DAA, direct acting antiviral; ALT, alanine aminotransferase; ULN, upper limit of normal; AST, aspartate aminotransferase.
*As assessed by the investigator.
†Two patients experienced a serious AE of angioedema leading to premature G/P discontinuation on Days 8 and 15, respectively.
‡Post-nadir increase in grade to Grade ≥3.

LIMITATIONS

- This was a single-arm, open-label trial without a placebo arm; however, the use of objective measures for efficacy (SVR12) and the comparison with a pre-specified threshold considering historical reference SVR12 rates (efficacy observed in the registrational program) mitigates this concern

CONCLUSIONS

- The study met its primary and secondary efficacy endpoints with SVR12 rates of 100% and 96.5% in mITT and ITT populations, respectively
 - No virologic failures were observed in treatment-naïve HCV GT1–6-infected patients with an APRI ≤1 treated with 8-week G/P regimen
- G/P was well-tolerated with no Grade ≥3 laboratory abnormalities in ALT, AST, and total bilirubin, as well as with low rates of serious AEs (2%) and AEs leading to premature G/P discontinuation (1%)
- Treating patients infected with HCV GTs 1–6 and with an APRI score of ≤1 with 8 weeks of G/P was highly efficacious and well tolerated
- Patients with APRI ≤1 and no prior evidence of cirrhosis can be successfully treated with G/P for 8 weeks, suggesting that APRI can be used in simplified HCV management strategies

PRESENTATION NOTES

Data to be presented at AASLD 2018 on EXPEDITION-8 (LB-7) that assessed safety and efficacy of 8-week G/P regimen in treatment-naïve patients with compensated cirrhosis and HCV GT1, 2, 4–6 infection.

ACKNOWLEDGEMENTS

- The authors would like to express their gratitude to the patients and their families, investigators, and coordinators who made this study possible.
- Medical writing support was provided by Dan O'Brien, PhD, of AbbVie.

DISCLOSURES

Robert J Fontana: Research support from BMS, Gilead, AbbVie; Consulting: Alynam. **Stuart McPherson:** Consultancy/speakers fees: AbbVie, Allergan, BMS, Gilead, MSD, Novartis. **Sabela Lens:** Advisor: Janssen, Gilead, AbbVie. **Magdy Elkhashab:** Grant support from AbbVie; Advisor for AbbVie. **Stefan Zeuzem:** Consultancies for AbbVie, BMS, Gilead, Janssen, Merck. **Krum Katzarov:** nothing to disclose. **Ana Gabriela Pires dos Santos, Roger Trinh, Zhenyi Xue, Lino Rodrigues Junior,** and **Ariel Porcalla** are employees of AbbVie Inc. and may hold stock or stock options.

AbbVie sponsored the study (NCT03212521), contributed to its design and collection, analysis, and interpretation of the data, and participated in the writing, review, and approval of the abstract. All authors had access to relevant data. Glecaprevir was identified by AbbVie and Enanta.

REFERENCES

1. World Health Organization. Global Hepatitis Report. Geneva, Switzerland. 2017.
2. Calvaruso V, et al. Is global elimination of HCV realistic? *Liver International*. 2018;38 (Suppl. 1):40–6.
3. Chou R and Wasson N. Blood tests to diagnose fibrosis or cirrhosis in patients with chronic hepatitis C virus infection. *Annals of Internal Medicine*. 2018;158(11):807–20.
4. Kelly ML, et al. Capacity of non-invasive hepatic fibrosis algorithms to replace transient elastography to exclude cirrhosis in people with hepatitis C virus infection: A multi-centre observational study. *PLoS ONE*. 2018;13:e0192763.
5. World Health Organization. Guidelines for the care and treatment of persons diagnosed with chronic hepatitis C infection. Geneva, Switzerland. 2018.
6. European Association for the Study of the Liver. EASL Recommendations on Treatment of Hepatitis C 2018. *J Hepatol*. 2018;69:461–511.
7. Hepatitis C Virus Infection Consensus Statement Working Group. Australian recommendations for the management of hepatitis C virus infection: a consensus statement (September 2018). Melbourne: Gastroenterological Society of Australia, 2018.
8. Shah H, et al. The management of chronic hepatitis C: 2018 guideline update from the Canadian Association for the Study of the Liver. *CMAJ*. 2018;190(22):E677–87.
9. MAVYRET [US Package Insert]/MAVIRET [SmPC], AbbVie 2017.
10. T Berg, U Naumann, A Stoehr, et al. GS-007, EASL/ILC, April 13, 2018.
11. R D'Ambrosio, L Pasulo, M Puoti, et al. GS-013, EASL/ILC, April 14, 2018.
12. Kamil RJ, et al. Case-control study evaluating competing risk factors for angioedema in a high-risk population. *Laryngoscope*. 2016;126(8):1823–30.

Scan QR code to download an electronic version of this presentation (poster 653) and other AbbVie AASLD 2018 Scientific Presentations. To obtain a QR code reader, go to your device app store and search for "QR code reader." QR code expiration: 27 November 2018.

abbvie

44073-HCV-FRI-653 — AASLD 2018 — AbbVie — proof 4 (rjh) — october 26, 2018
the henderson company 6020 keating avenue, chicago illinois 60646 (847) 979-8051

CREATED @ 100%